Survodutide, a glucagon receptor/GLP-1 receptor (GCGR/GLP-1R) dual agonist, improves cardiometabolic parameters in adults with obesity: Analysis of a placebo-controlled, randomized phase 2 trial

Carel W. le Roux,[1] Oren Steen,[2] Kathryn J. Lucas,[3] Elif I. Ekinci,[4] Elena Startseva,[5] Anna Unseld,[6] Anita M. Hennige[7]

[1]St. Vincent's University Hospital and University College Dublin School of Medicine, Dublin, Ireland; [2]Private practice, Toronto, Ontario, Canada; [3]Diabetes & Endocrinology Consultants PC, Morehead City, NC, USA; [4]Austin Health, Heidelberg, Victoria, Australia; The Australian Centre for Accelerating Diabetes Innovation, University of Melbourne, Parkville, Australia; and Melbourne Medical School, University of Melbourne, Melbourne, Victoria, Australia; [5]Boehringer Ingelheim International GmbH, Ingelheim, Germany; [6]Boehringer Ingelheim Pharma GmbH & Co. KG, Biberach, Germany; [7]Boehringer Ingelheim International GmbH, Biberach, Germany

Objective

- To further analyze the phase 2 trial of survodutide in people living with overweight or obesity (BMI ≥27 kg/m²), to explore:
 - The effect of survodutide on cardiometabolic parameters in this trial cohort

Conclusions

- Survodutide was associated with clinically meaningful improvements in cardiometabolic parameters in people living with overweight or obesity
- Reductions were observed in BP (regardless of hypertension status at baseline), waist circumference, and plasma TG

Scan the QR code or visit the URL to view the e-poster https://go.boehringer.com/qwOI

Introduction

- Obesity increases the risk of cardiovascular disease and contributes to cardiovascular risk factors, such as hypertension and dyslipidemia.[1] The risk of cardiometabolic disease is reduced following sustained body weight reduction in individuals with obesity[1]
- Combining GCGR agonism with GLP-1R agonism may enhance body weight reduction by increasing energy expenditure, as well as decreasing food intake[2]
- Survodutide (BI 456906) is a GCGR/GLP-1R dual agonist under investigation for chronic weight management in people living with obesity[3]
- In a multinational phase 2 trial in individuals with overweight or obesity and without diabetes (NCT04667377), survodutide elicited up to 18.7% mean reduction in body weight after 46 weeks of treatment (according to actual maintenance doses)[4]

Methods

- The trial design is shown in **Figure 1**.[4] Here, changes from baseline to Week 46 were evaluated for BP (by presence/absence of hypertension before and at screening), waist circumference, and lipid parameters (TG, HDL, VLDL, LDL, TC, and non–HDL-C)
- Data were analyzed for all participants receiving ≥1 dose of study drug with data for ≥1 efficacy endpoint (i.e. FAS), according to doses received during the maintenance period (actual treatment), and according to doses assigned at randomization (planned treatment) using on-treatment data
- MMRM was used to generate estimates of mean absolute changes from baseline in SBP, DBP, and waist circumference at Week 46 for each dose group

Figure 1. Trial design

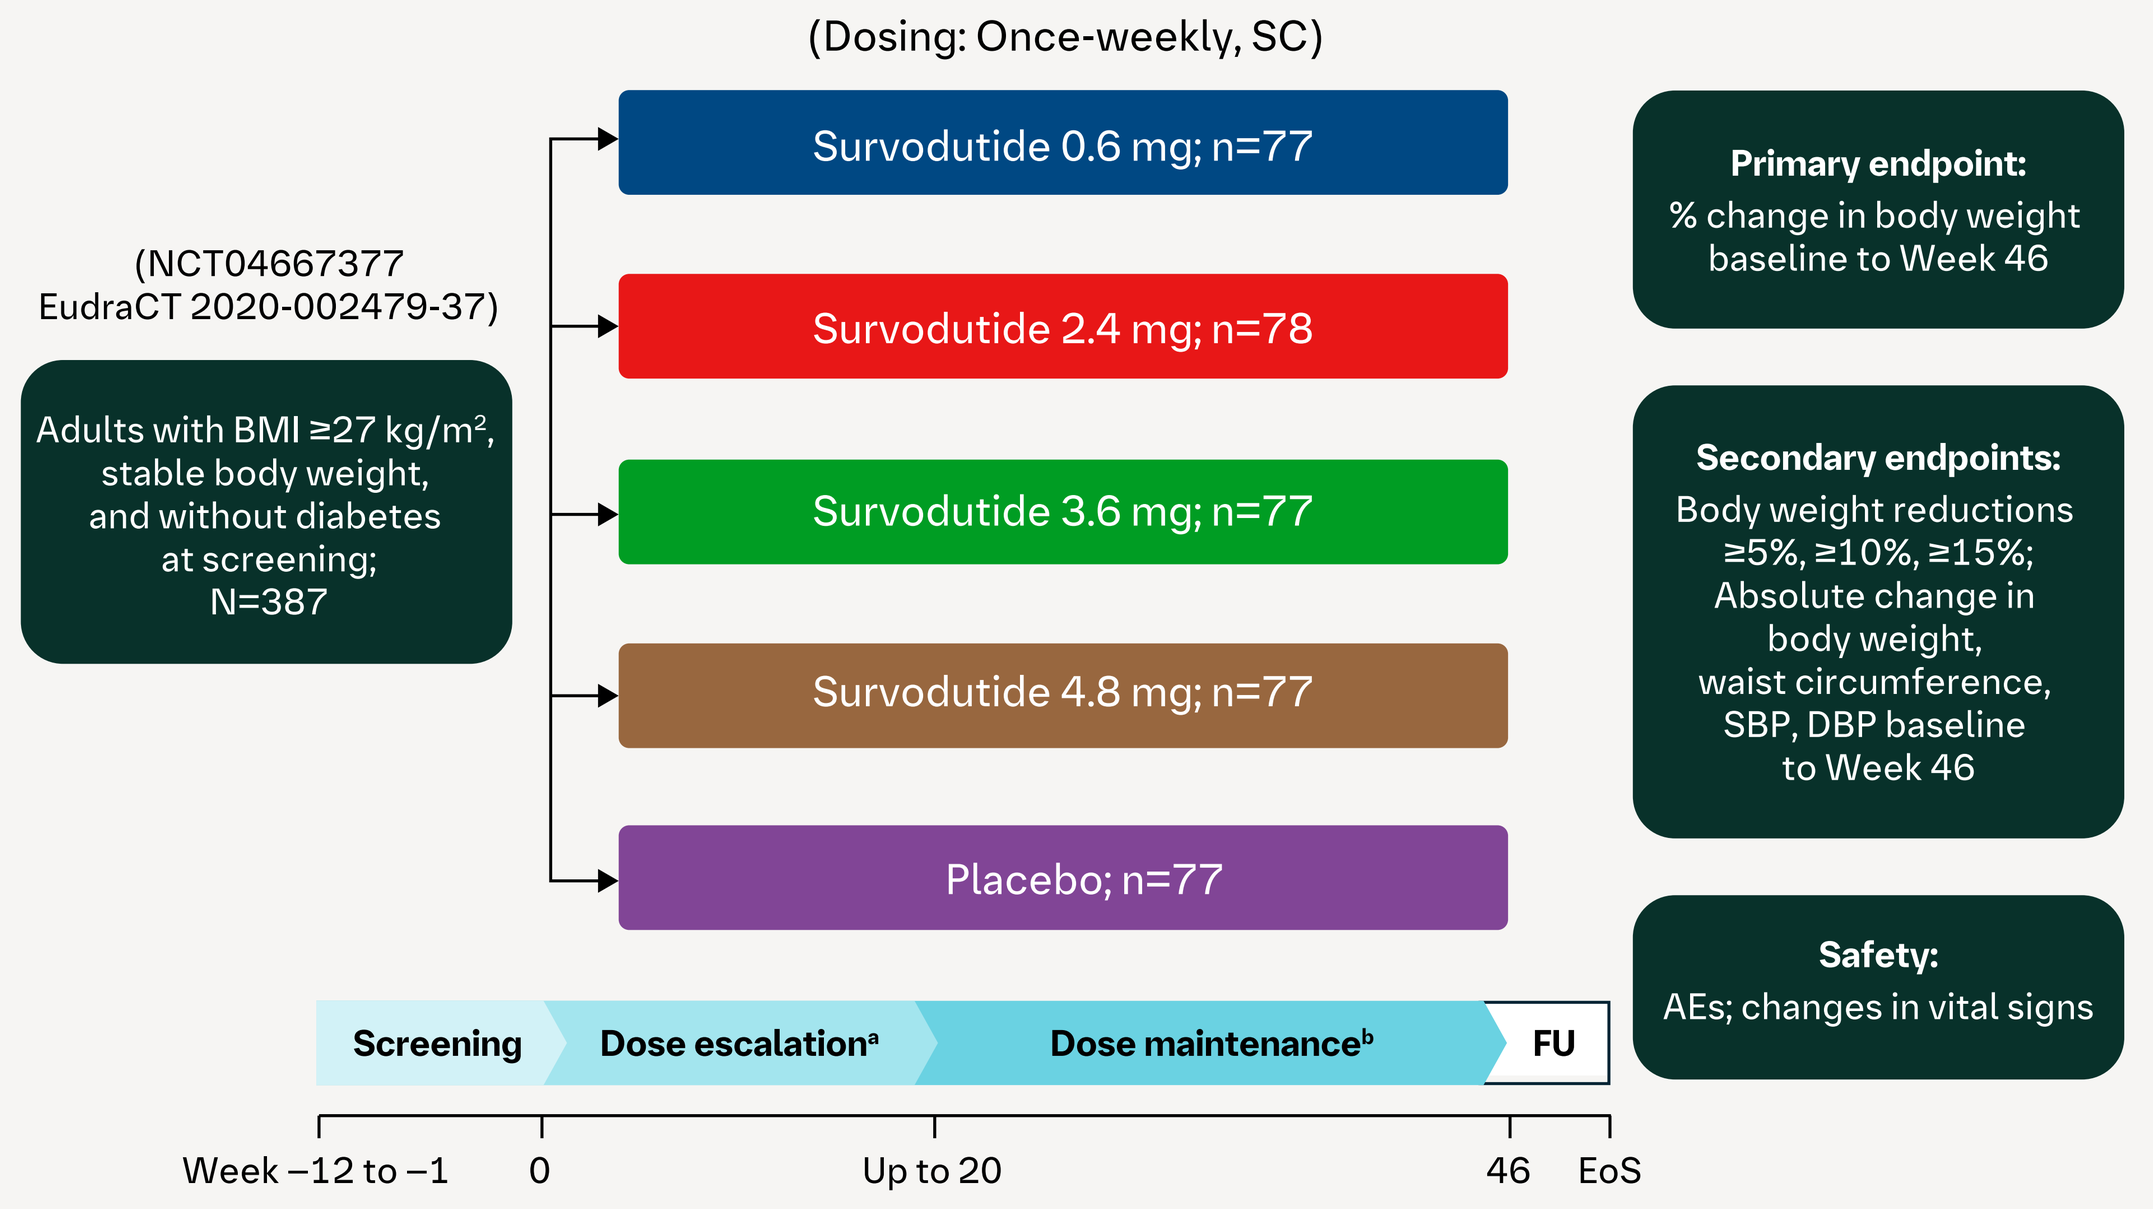

(Dosing: Once-weekly, SC)
(NCT04667377 EudraCT 2020-002479-37)
Adults with BMI ≥27 kg/m², stable body weight, and without diabetes at screening; N=387
Survodutide 0.6 mg; n=77
Survodutide 2.4 mg; n=78
Survodutide 3.6 mg; n=77
Survodutide 4.8 mg; n=77
Placebo; n=77
Primary endpoint: % change in body weight baseline to Week 46
Secondary endpoints: Body weight reductions ≥5%, ≥10%, ≥15%; Absolute change in body weight, waist circumference, SBP, DBP baseline to Week 46
Safety: AEs; changes in vital signs
Screening
Dose escalation[a]
Dose maintenance[b]
FU
Week −12 to −1
0
Up to 20
46
EoS

[a]In Weeks 11–20, tolerability was assessed every 2 weeks; if GI AEs were intolerable, the participant remained on the same dose for another week prior to dose escalation. [b]Participants who did not tolerate treatment due to GI AEs during dose escalation could remain on a lower survodutide dose (than the dose allocated to them at randomization) for the duration of the maintenance phase. Participants who could not tolerate the lowest survodutide dose tested (0.6 mg) despite all the efforts taken discontinued treatment.

Results

- In the FAS (n=384), baseline demographics and clinical characteristics were similar across treatment groups
 - 262 (68.2%) female, 301 (78.4%) White, 40 (10.4%) Asian, and 37 (9.6%) Black; overall mean age 49.1 years, BMI 37.1 kg/m², weight 105.7 kg, waist circumference 113.4 cm, SBP 125.6 mmHg, DBP 81.3 mmHg. At baseline, 133 (34.6%) participants had hypertension and 108 (28.1%) had dyslipidemia

BP parameters: change from baseline to Week 46

- Reductions in mean SBP (and DBP; data not shown) at Week 46 were observed in all survodutide dose groups tested; similar results were observed by planned and actual treatment (**Figure 2**)

Figure 2. MMRM estimates for change in SBP from baseline to Week 46 (FAS, on-treatment data)

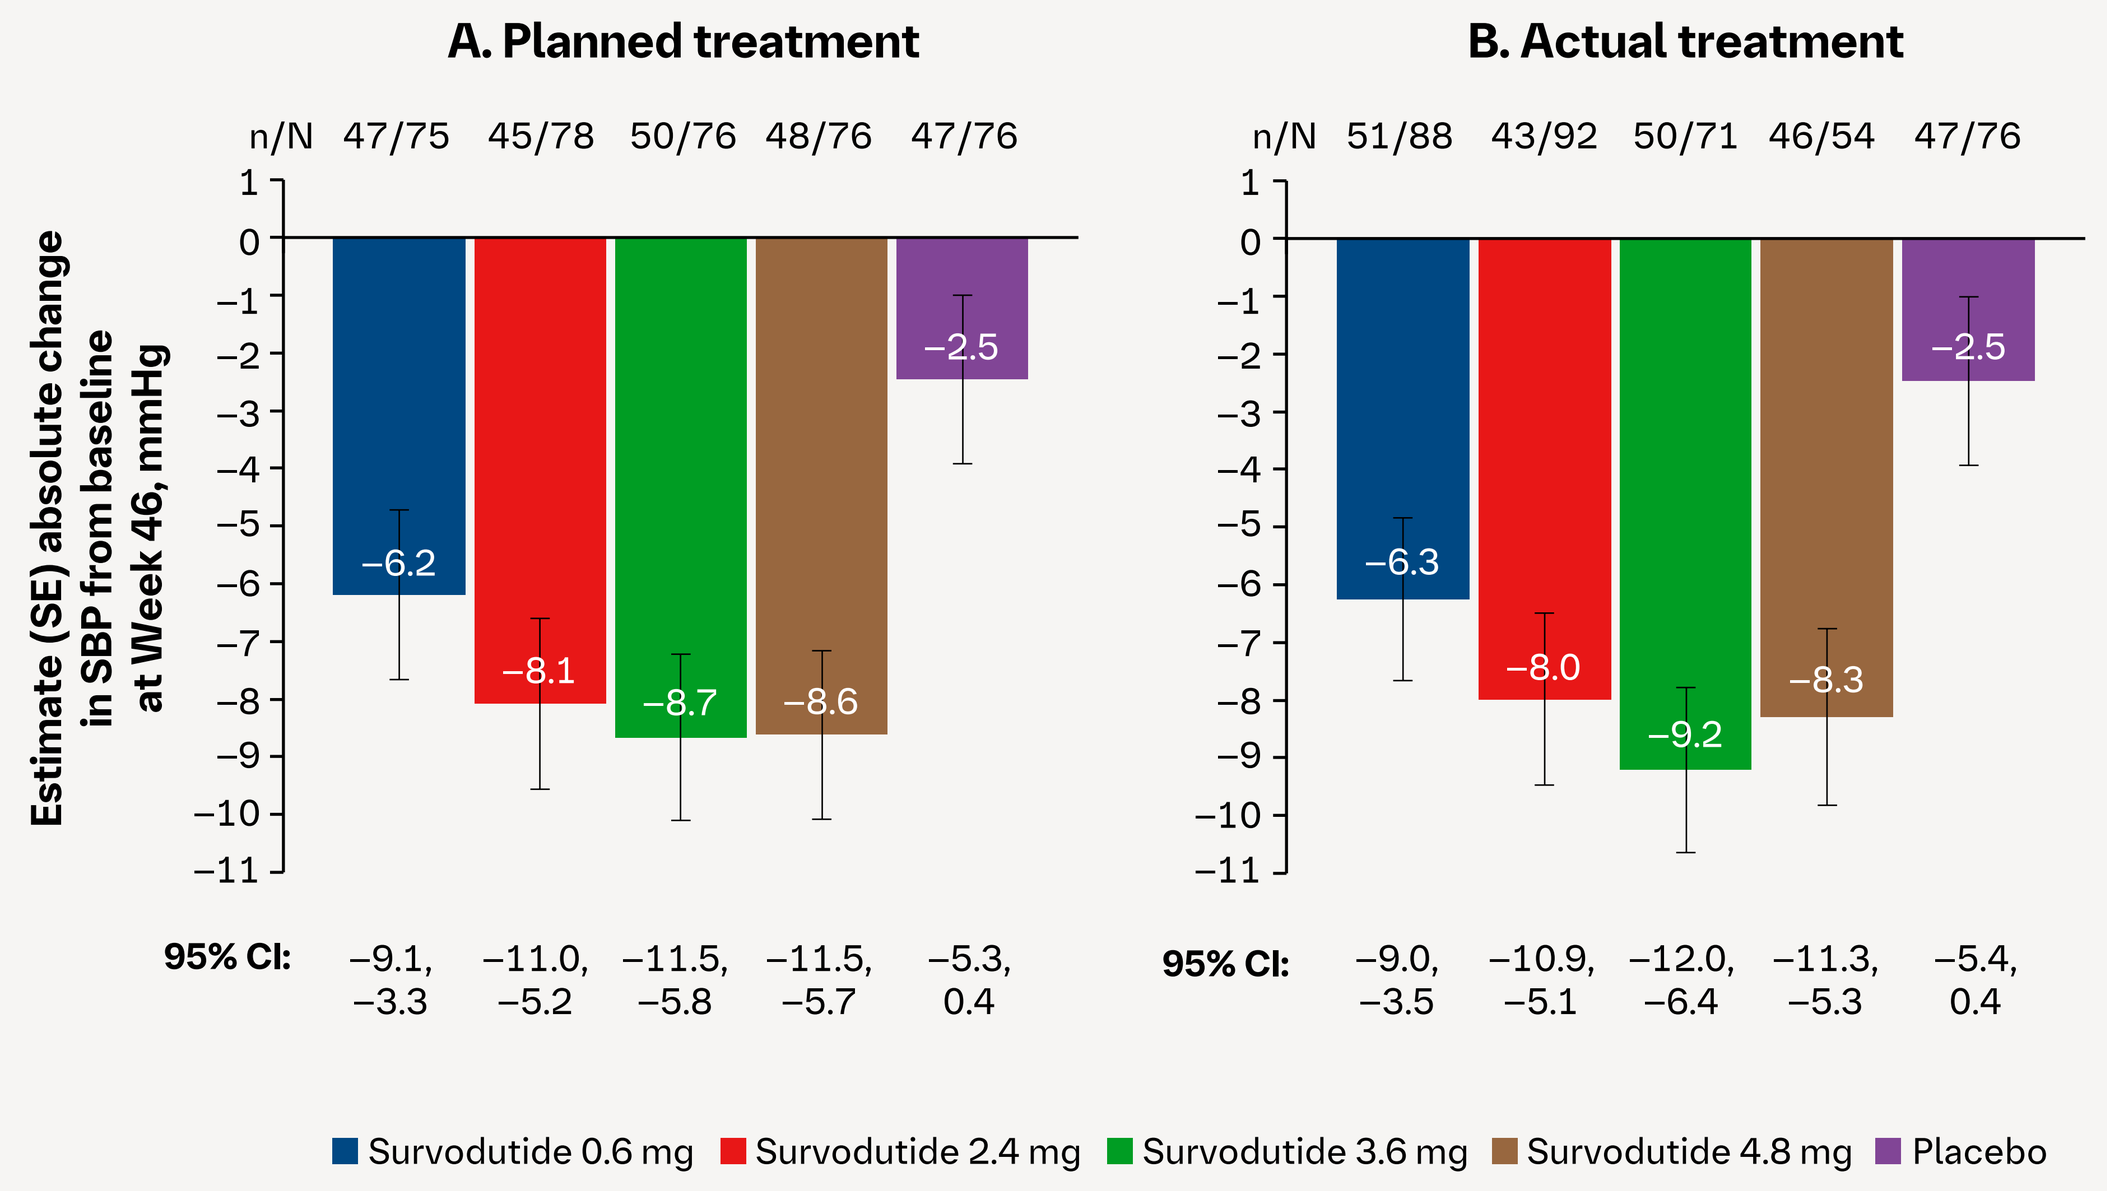

A. Planned treatment
n/N 47/75 45/78 50/76 48/76 47/76
Estimate (SE) absolute change in SBP from baseline at Week 46, mmHg
−6.2
−8.1
−8.7
−8.6
−2.5
95% CI: −9.1, −3.3; −11.0, −5.2; −11.5, −5.8; −11.5, −5.7; −5.3, 0.4
B. Actual treatment
n/N 51/88 43/92 50/71 46/54 47/76
−6.3
−8.0
−9.2
−8.3
−2.5
95% CI: −9.0, −3.5; −10.9, −5.1; −12.0, −6.4; −11.3, −5.3; −5.4, 0.4
Survodutide 0.6 mg
Survodutide 2.4 mg
Survodutide 3.6 mg
Survodutide 4.8 mg
Placebo

Numbers per dose group are given as n at Week 46/N at baseline.

- Reductions in SBP (and DBP; data not shown) were comparable when participants were analyzed according to the presence or absence of hypertension at baseline (**Figure 3**). SBP reductions were slightly larger in participants who were normotensive at baseline

Figure 3. Change in SBP from baseline to Week 46 by hypertension status at baseline[a] (FAS, on-treatment data)

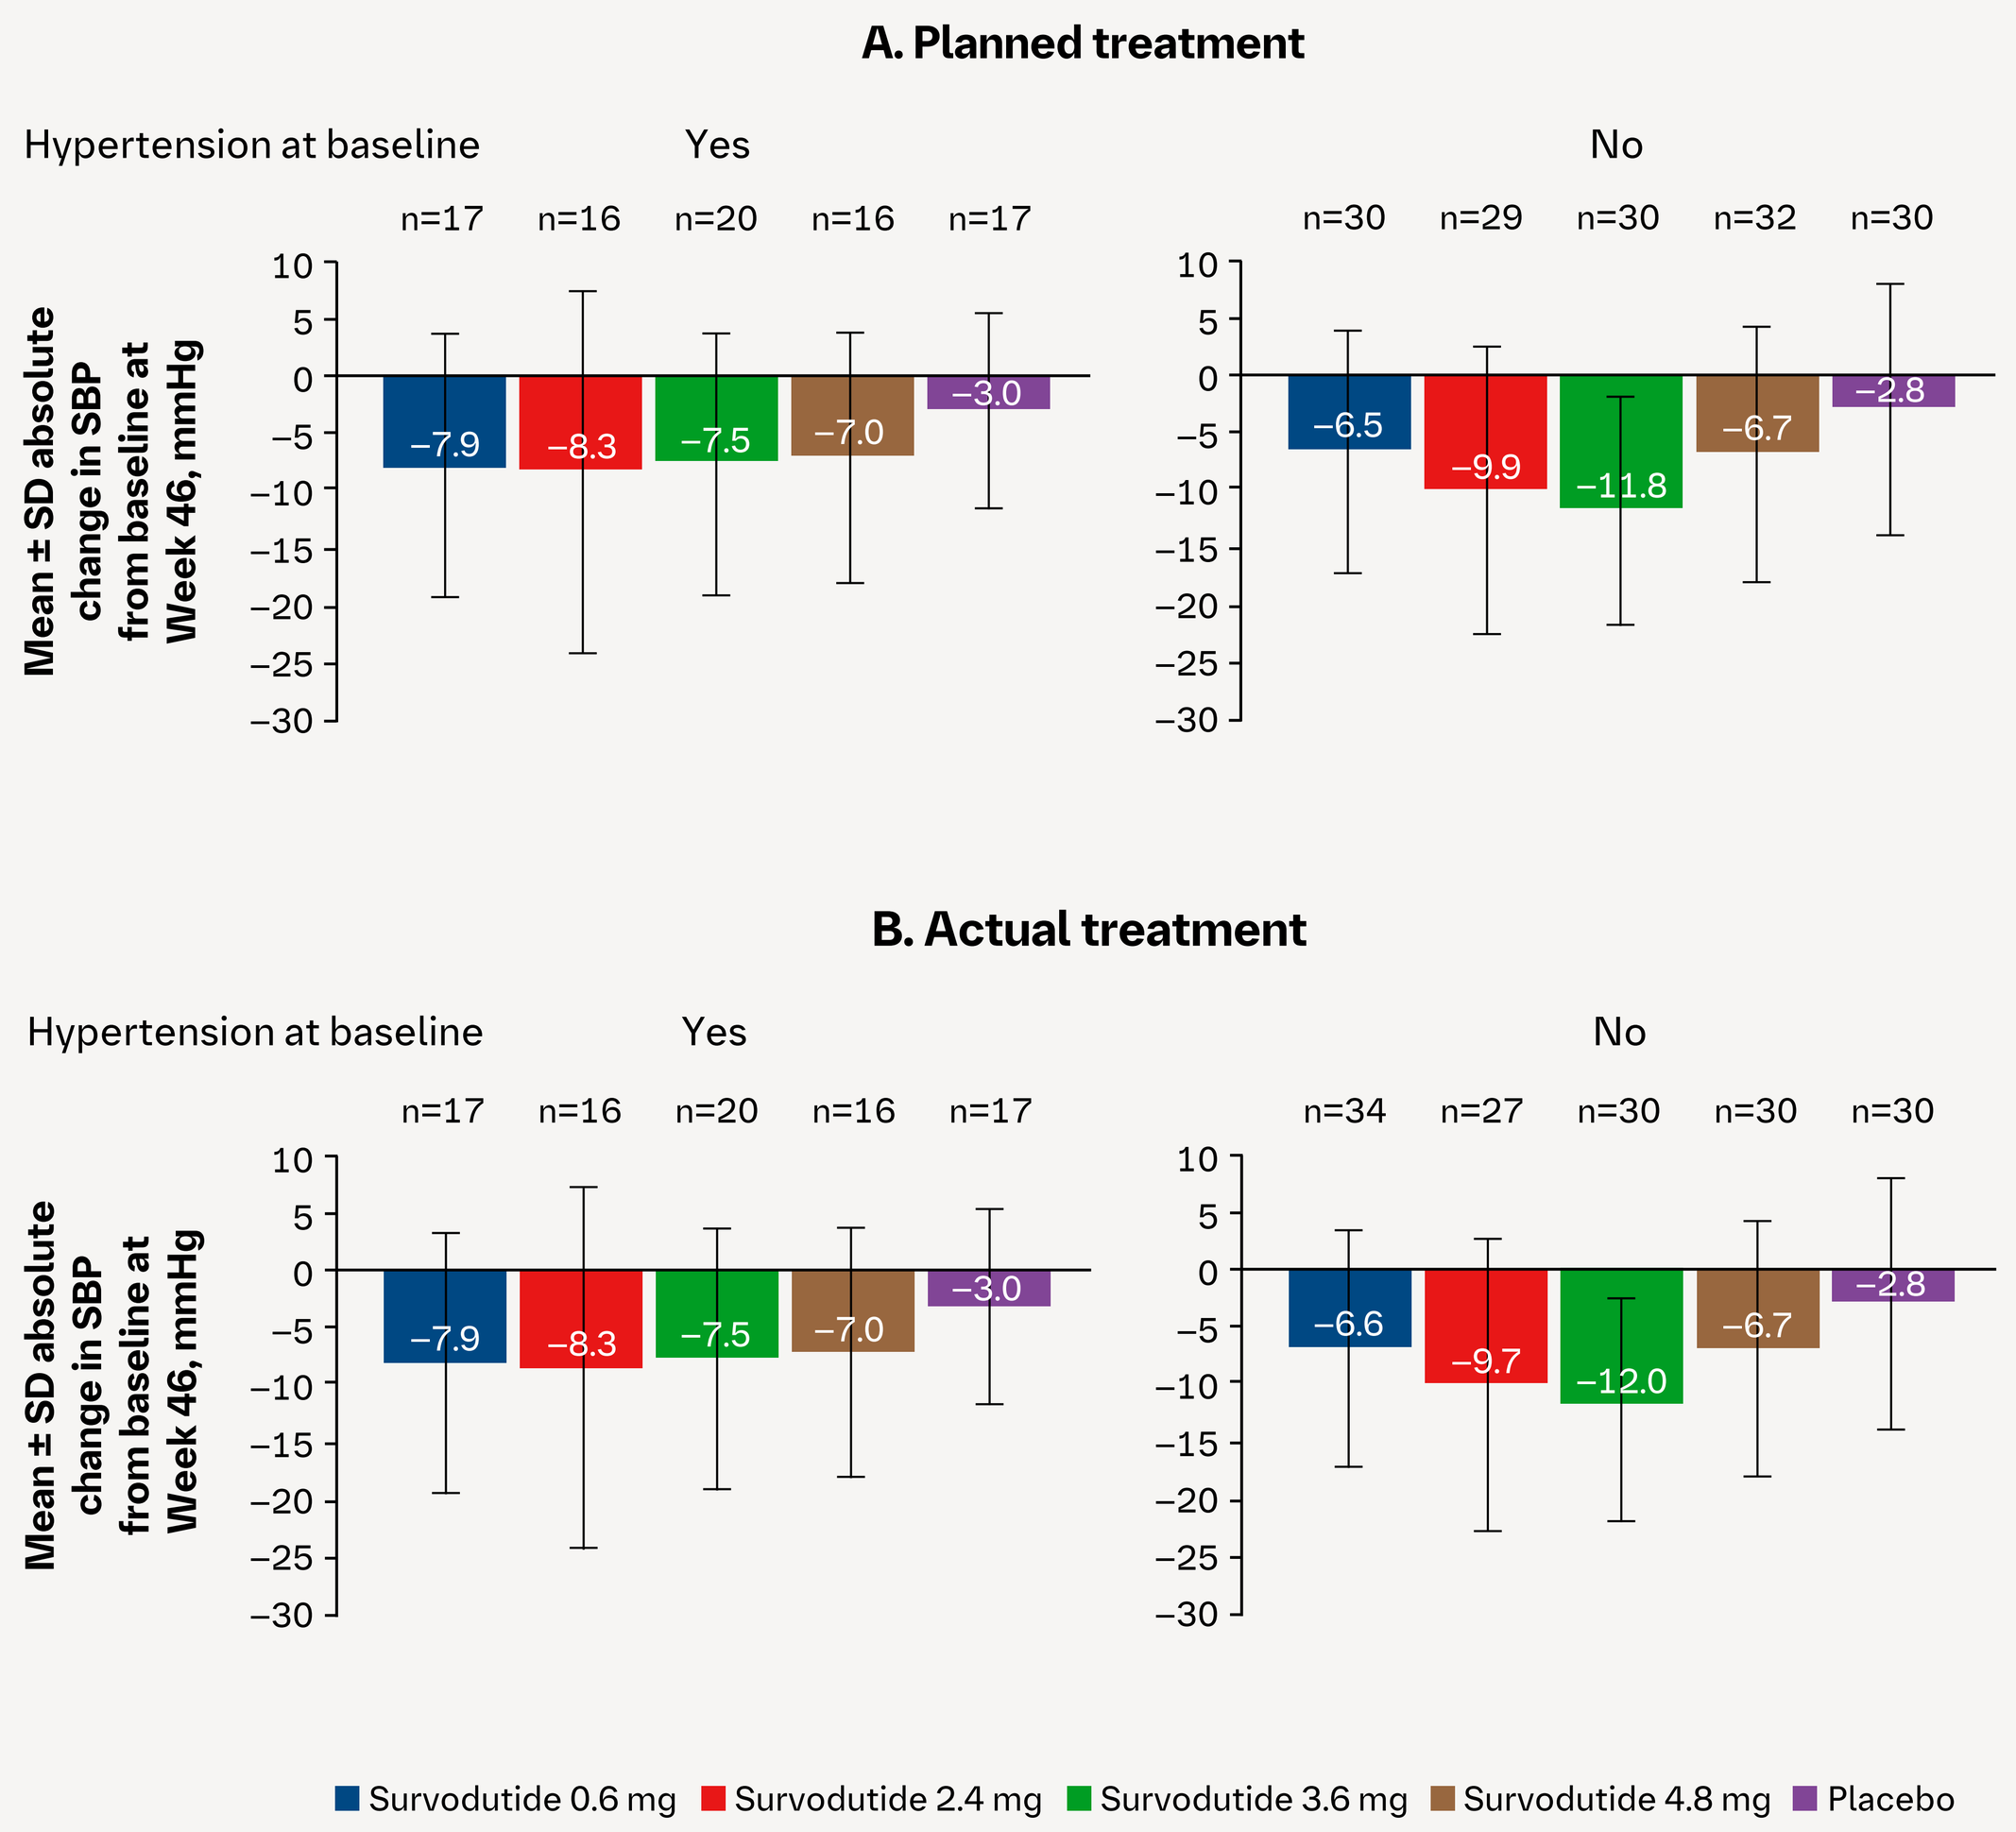

A. Planned treatment
Hypertension at baseline
Yes
n=17 n=16 n=20 n=16 n=17
−7.9
−8.3
−7.5
−7.0
−3.0
No
n=30 n=29 n=30 n=32 n=30
−6.5
−9.9
−11.8
−6.7
−2.8
Mean ± SD absolute change in SBP from baseline at Week 46, mmHg
B. Actual treatment
Hypertension at baseline
Yes
n=17 n=16 n=20 n=16 n=17
−7.9
−8.3
−7.5
−7.0
−3.0
No
n=34 n=27 n=30 n=30 n=30
−6.6
−9.7
−12.0
−6.7
−2.8
Survodutide 0.6 mg
Survodutide 2.4 mg
Survodutide 3.6 mg
Survodutide 4.8 mg
Placebo

Descriptive statistics are presented. [a]Defined as: before and at screening.

Safety

- Dyslipidemia: 2 AEs with survodutide (n=1 each for 2.4 mg and 3.6 mg dose groups); 1 AE with placebo; all AEs were mild, non-serious, and did not result in discontinuation of study medication
- Hypotension: 5 AEs with survodutide (n=1 with underlying hypertension); 1 AE with placebo (with underlying hypertension). Orthostatic hypotension: 2 AEs with survodutide. All AEs were non-serious, but 1 AE was severe (participant in survodutide 4.8 mg-dose group with underlying hypertension)
 - In participants who were normotensive at baseline, SBP reductions were not associated with any serious or severe hypotensive episodes

Waist circumference: change from baseline to Week 46

- Mean waist circumference at Week 46 was lower in all survodutide dose groups tested. The reductions were of a similar magnitude when participants were analyzed by planned and actual treatment received (**Figure 4**)

Figure 4. MMRM estimates for change in waist circumference from baseline to Week 46 (FAS, on-treatment data)

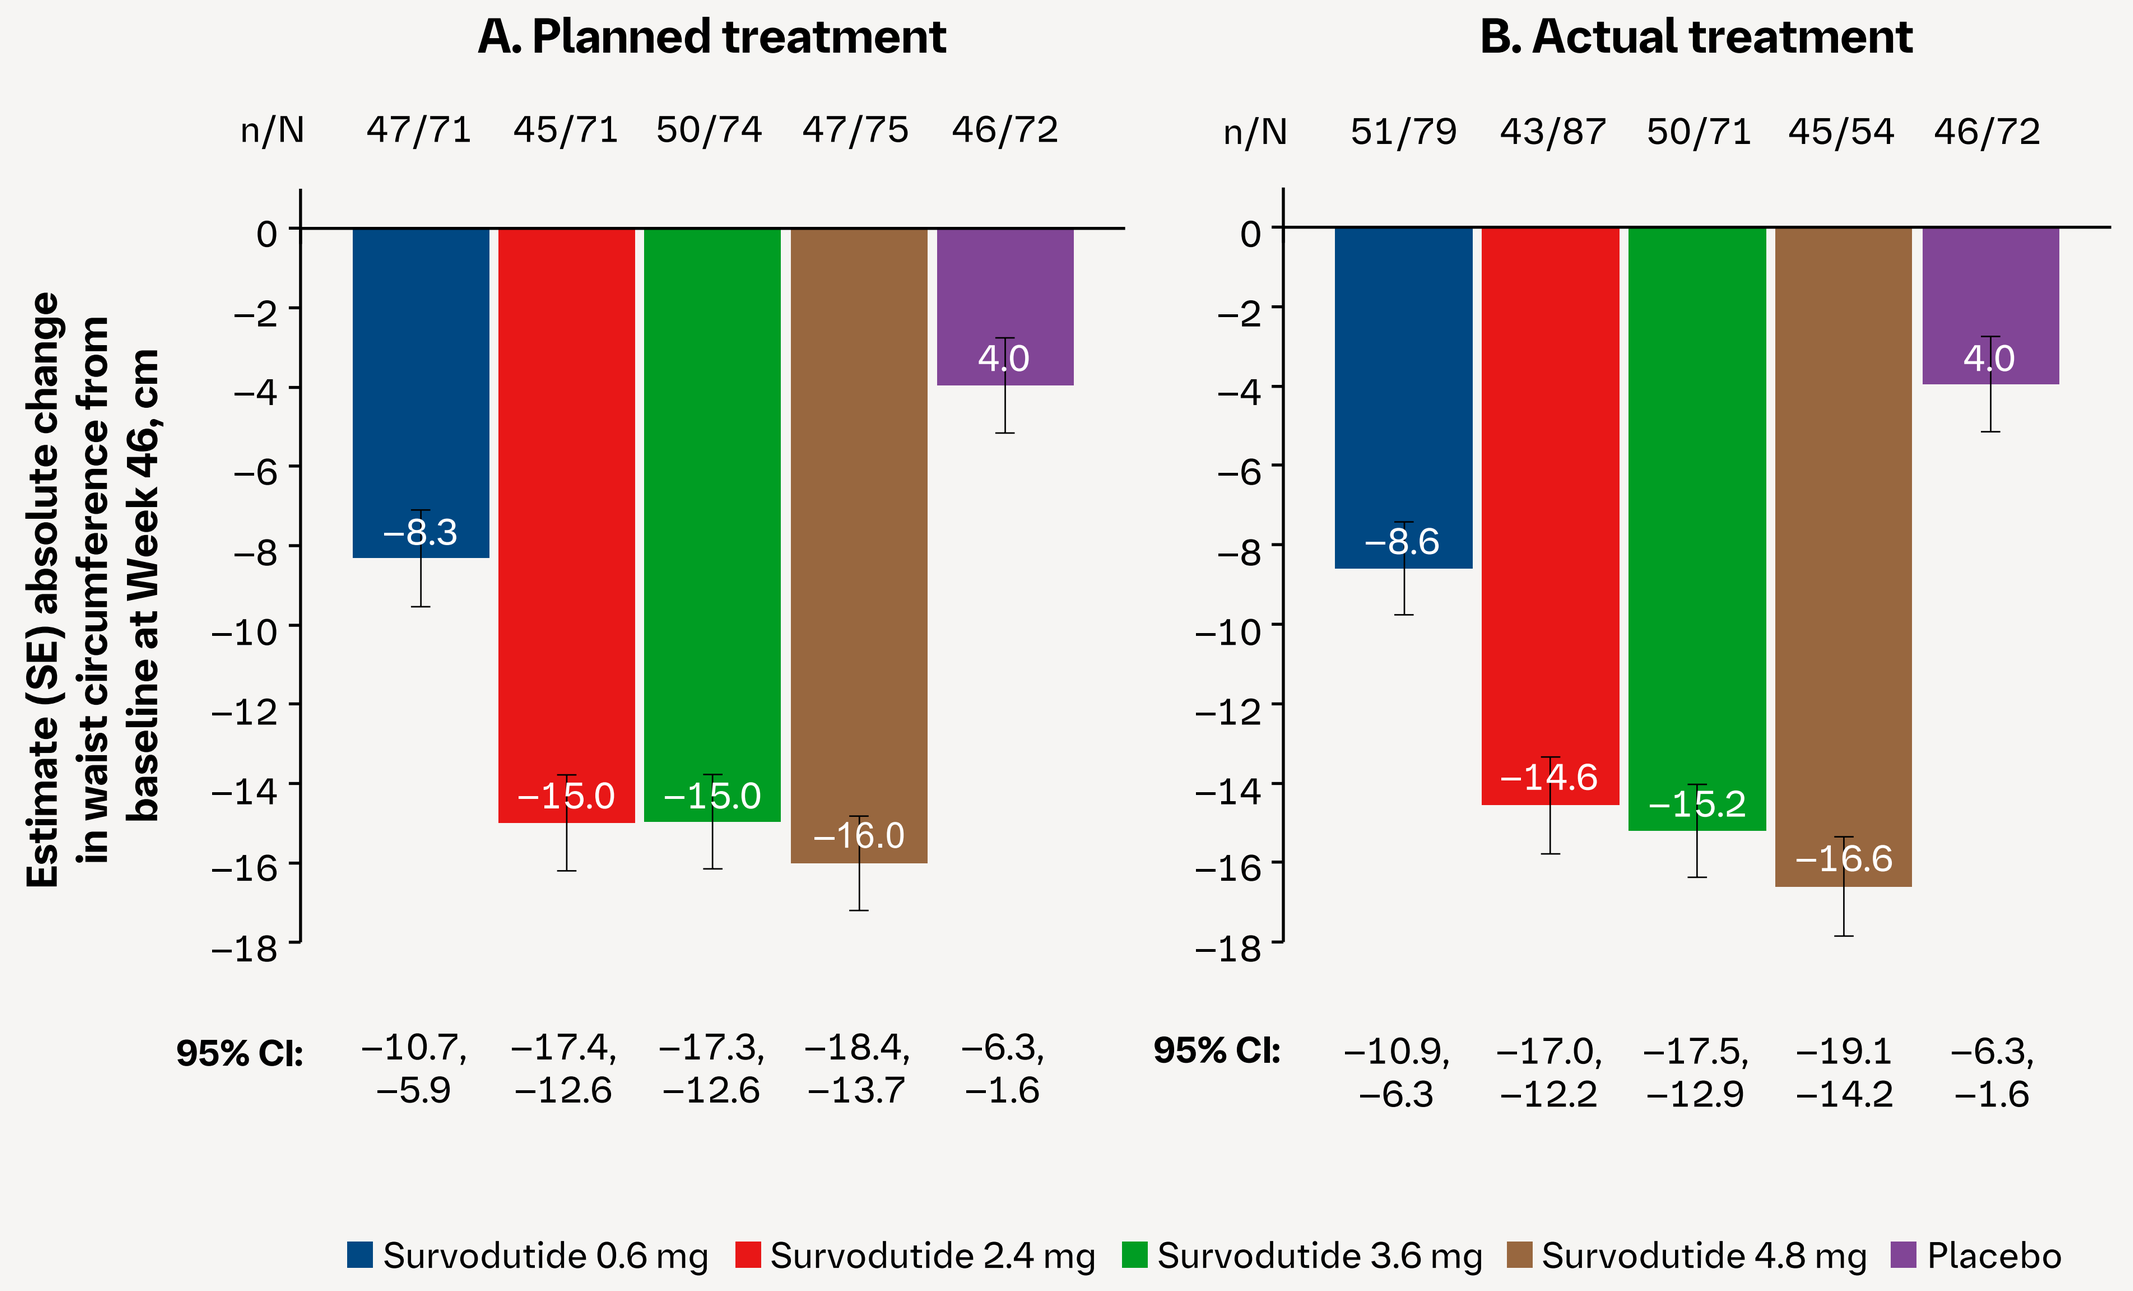

A. Planned treatment
n/N 47/71 45/71 50/74 47/75 46/72
Estimate (SE) absolute change in waist circumference from baseline at Week 46, cm
−8.3
−15.0
−15.0
−16.0
4.0
95% CI: −10.7, −5.9; −17.4, −12.6; −17.3, −12.6; −18.4, −13.7; −6.3, −1.6
B. Actual treatment
n/N 51/79 43/87 50/71 45/54 46/72
−8.6
−14.6
−15.2
−16.6
4.0
95% CI: −10.9, −6.3; −17.0, −12.2; −17.5, −12.9; −19.1, −14.2; −6.3, −1.6
Survodutide 0.6 mg
Survodutide 2.4 mg
Survodutide 3.6 mg
Survodutide 4.8 mg
Placebo

Numbers per dose group are given as n at Week 46/N at baseline.

Lipid parameters: change from baseline to Week 46

- Mean plasma TG concentrations were decreased in all survodutide dose groups analyzed according to planned treatment (**Table 1**). Mean HDL concentrations were relatively unchanged
- VLDL concentrations were lower in participants receiving survodutide, but not in those receiving placebo. There was no consistent pattern to the observed changes in plasma levels of LDL, TC, and non–HDL-C (data not shown)
- The proportion of participants with and without dyslipidemia at Week 46 was largely unchanged from baseline

Table 1. Change in lipid parameters from baseline to Week 46 (FAS, planned treatment, on-treatment data)

Change from baseline, mean (± SD)	Survodutide 0.6 mg (n=77)[a]	Survodutide 2.4 mg (n=78)[a]	Survodutide 3.6 mg (n=76)[a]	Survodutide 4.8 mg (n=76)[a]	Placebo (n=77)[a]
TG					
Baseline, mg/dL	147.6 (± 79.6)	143.3 (± 78.2)	137.4 (± 62.6)	134.6 (± 56.6)	115.8 (± 52.0)
Absolute change, mg/dL	−31.1 (± 49.8) n=46	−46.9 (± 52.4) n=45	−50.4 (± 62.4) n=50	−30.7 (± 58.3) n=48	−1.6 (± 40.7) n=46
% change	−16.7 (± 26.6) n=46	−27.3 (± 22.8) n=45	−27.7 (± 24.9) n=50	−19.9 (± 30.8) n=48	5.7 (± 36.4) n=46
Baseline, mmol/L	1.7 (± 0.9)	1.6 (± 0.9)	1.6 (± 0.7)	1.5 (± 0.6)	1.3 (± 0.6)
Absolute change, mmol/L	−0.35 (± 0.56)	−0.53 (± 0.59)	−0.57 (± 0.70)	−0.35 (± 0.66)	−0.02 (± 0.46)
HDL					
Baseline, mg/dL	51.3 (± 12.8)	50.9 (± 14.0)	50.4 (± 12.7)	54.0 (± 14.6)	54.1 (± 13.5)
Absolute change, mg/dL	−0.6 (± 6.9) n=46	2.9 (± 8.7) n=45	−0.5 (± 6.4) n=50	−0.3 (± 9.1) n=48	0.1 (± 6.0) n=46
% change	0.5 (± 14.8) n=46	8.0 (± 17.4) n=45	0.2 (± 12.0) n=50	1.4 (± 17.2) n=48	0.3 (± 11.4) n=46
Baseline, mmol/L	1.3 (± 0.3)	1.3 (± 0.4)	1.3 (± 0.3)	1.4 (± 0.4)	1.4 (± 0.4)
Absolute change, mmol/L	−0.02 (± 0.18)	0.08 (± 0.23)	−0.01 (± 0.17)	−0.01 (± 0.24)	0.00 (± 0.16)
VLDL					
Baseline, mg/dL	29.0 (± 14.2)	27.6 (± 13.2) n=77	27.5 (± 12.5)	26.9 (± 11.3)	23.2 (± 10.4)
Absolute change, mg/dL	−6.4 (± 10.0) n=45	−8.3 (± 8.0) n=44	−10.1 (± 12.4) n=50	−6.2 (± 11.7) n=48	−0.4 (± 8.1) n=46
% change	−17.2 (± 26.6) n=45	−26.6 (± 22.4) n=44	−28.0 (± 24.6) n=50	−20.4 (± 31.1) n=48	5.5 (± 36.4) n=46
Baseline, mmol/L	0.8 (± 0.4)	0.7 (± 0.3)	0.7 (± 0.3)	0.7 (± 0.3)	0.6 (± 0.3)
Absolute change, mmol/L	−0.17 (± 0.26)	−0.21 (± 0.21)	−0.26 (± 0.32)	−0.16 (± 0.30)	−0.01 (± 0.21)

Descriptive statistics are presented. Data are presented in conventional (mg/dL) and SI units (mmol/L). Conversion of TG from mg/dL to mmol/L: value in mg/dL is multiplied by 0.01129 (or divided by 88.57); conversion of HDL and VLDL from mg/dL to mmol/L: value in mg/dL is multiplied by 0.02586 (or divided by 38.67).

[a]n stated if value differs from baseline.

Presented at: the 19th Annual Cardiometabolic Health Congress, Boston, MA, USA; October 17–19, 2024

Previously presented at: the European Society of Cardiology (ESC) Congress 2024, London, UK/Hybrid; Aug 30–Sept 2, 2024

Abbreviations

AE, adverse event; BMI, body mass index; BP, blood pressure; CI, confidence interval; DBP, diastolic blood pressure; EoS, end of study; FAS, full analysis set; FU, follow-up; GCGR, glucagon receptor; GI, gastrointestinal; GLP-1R, glucagon-like peptide-1 receptor; HDL, high-density lipoprotein; LDL, low-density lipoprotein; MMRM, mixed model for repeated measures; non–HDL-C, non–HDL cholesterol; SBP, systolic blood pressure; SC, subcutaneous; SD, standard deviation; SE, standard error; TC, total cholesterol; TG, triglyceride; VLDL, very low-density lipoprotein

References

1. Valenzuela PL, et al. *Nat Rev Cardiol.* 2023;20:475–94.
2. Del Prato S, et al. *Obesity Rev.* 2022;23:e13372.
3. Zimmermann T, et al. *Mol Metab.* 2022;66:101633.
4. le Roux CW, et al. *Lancet Diabetes Endocrinol.* 2024;12:162–73.

Disclosures

CWlR has received personal fees from Boehringer Ingelheim, Eli Lilly, GI Dynamics, Gila Pharmaceuticals, Herbalife, Johnson & Johnson, Keyron, Novo Nordisk, and Zealand Pharma outside the submitted work. **OS** has received research support from Alnylam, Anji, AstraZeneca, Boehringer Ingelheim, CRISPR Therapeutics, Eli Lilly, Gilead Sciences, Janssen, Kowa, Medicago, Moderna, Novartis, Novo Nordisk, Pfizer, Sanofi, ViaCyte, and Zucara Therapeutics; speaker bureau fees from Abbott, Amgen, AstraZeneca, Bausch Health, Boehringer Ingelheim, Eli Lilly, Janssen, LMC, Novo Nordisk, and Sanofi; and consultancy fees from Amgen, Bayer, Eli Lilly, Novo Nordisk, and Sanofi. **KJL** declares no conflicts of interest. **EIE** is a consultant for Eli Lilly Australia, and her institute reports research funding support from Amgen, Bayer, Boehringer Ingelheim, Eli Lilly Australia, and Versanis. **ES**, **AU** and **AMH** are employees of Boehringer Ingelheim.

Acknowledgments

Survodutide is licensed to Boehringer Ingelheim (BI) from Zealand Pharma, with BI solely responsible for development and commercialization globally. Zealand has a co-promotion right in the Nordic countries. The authors thank the study participants, investigators, and study site staff. The study was supported and funded by BI. The authors met criteria for authorship as recommended by the International Committee of Medical Journal Editors (ICMJE). The authors did not receive payment related to the development of this poster. BI was given the opportunity to review the poster for medical and scientific accuracy as well as intellectual property considerations. Debra Brocksmith, MB ChB, PhD, of Elevate Scientific Solutions LLC (a member of Envision Pharma Group), provided medical writing support, which was contracted and funded by BI.

Boehringer Ingelheim